Neural correlates of individual differences in generous preferences

A.S. Fox[1,2,3,4,5*], N.J. Vack[2], A.A. Freeman[1] R.J. Davidson[1,2,3,4,5]
[1]Center for Investigating Healthy Minds, [2]Waisman Laboratory for Brain Imaging and Behavior, [3]Health Emotions Research Institute, and Departments of [4]Psychology and [5]Psychiatry at the University of Wisconsin Madison.
*Correspondence at: asfox@wisc.edu

Center for Investigating Healthy Minds

WISCONSIN
UNIVERSITY OF WISCONSIN–MADISON

Task

You get to choose. Either you get $1 or a stranger gets $5. What do you choose?

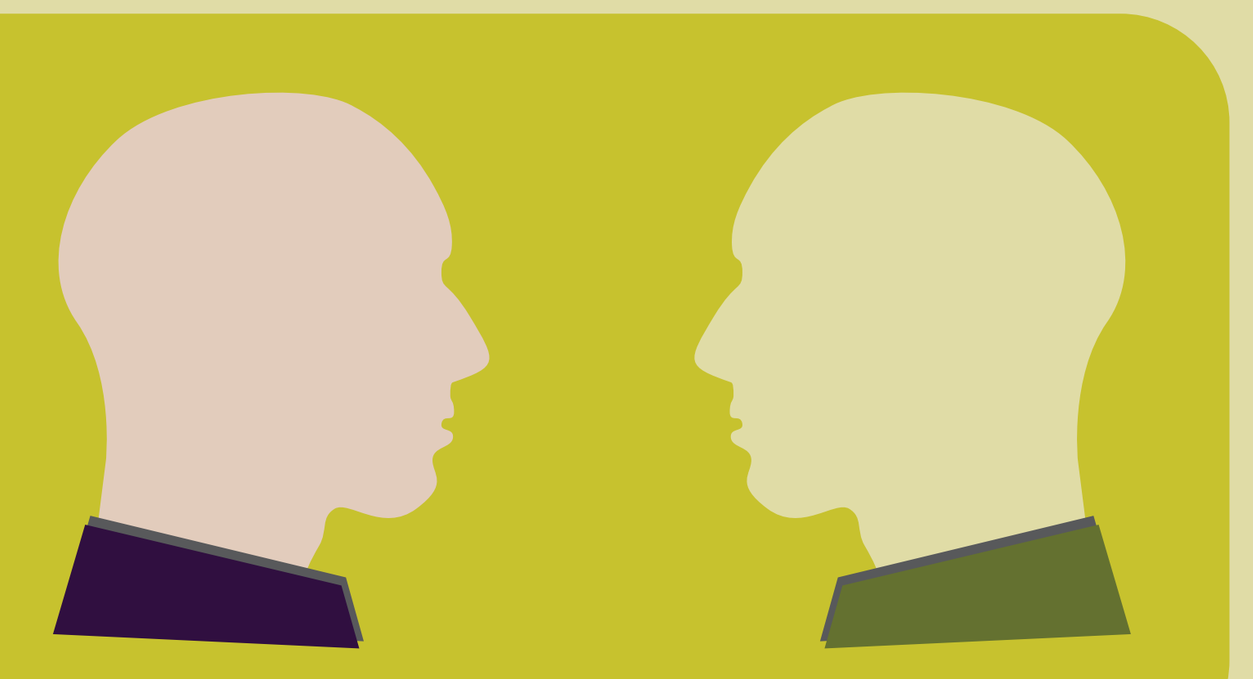

In this revealed prosocial preference task (RPP) we used real money and no deception. All interactions were anonymous, and there was no oppotunity for reciprocity. Any subjects who self-reported disbelief were excluded.

Estimation Technique

Step 1: Participant makes a choice.

We gave subjects 200 forced choice monetary decisions between money for themselves ($0-$10) and money for a stranger ($0-$10). To precisely quantify the amount of money an individual was willing to sacrifice to give a dollar to a stranger (i.e. their indifference point or "IP"), we adaptively altered the ratio of the two offers.

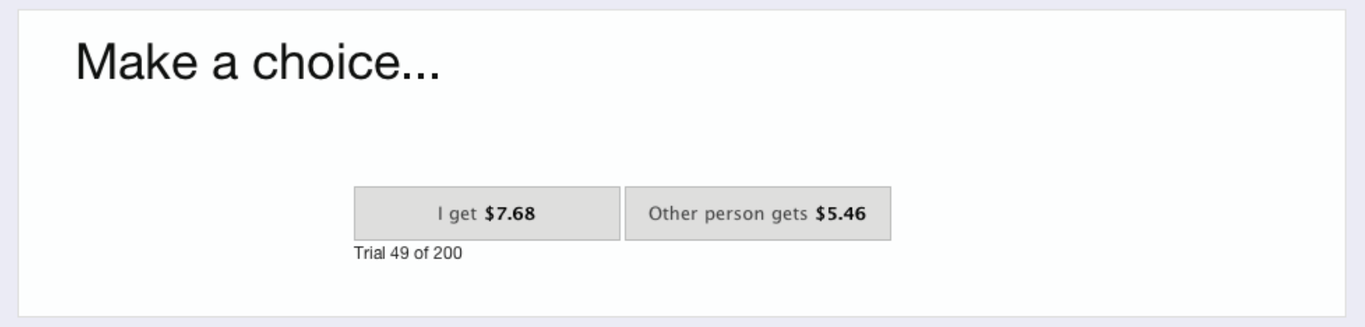
Make a choice...
I get $7.68
Other person gets $5.46
Trial 49 of 200

Step 2: Estimate an indifference point (IP).

$$\mathbf{C}_{[\mathbf{1:t}]} = \frac{1}{1+e^{\left(\alpha+\beta\times\left(\ln\left(OR_{[1:t]}\right)\right)\right)}}$$

$C_{[1:t]}$ is a vector of indicator variables {0,1} for each choice from the first trial through the current trial (t), where ones indicate choices for self. OR is the offer ratio for trial t. Using this model, we estimate α & β. α is the parameter estimate to a constant. β is the parameter estimate of interest, and represents the weight applied to the offer ratio. Using these parameters, we estimate indifference points from ln(-3) to ln(3).

$$\hat{\mathbf{IP}} = \frac{-\alpha}{\beta}$$

Step 3: Pick a ratio based on a normal distribution centered around the participant's indifference point (IP).

$$\mathbf{OR_{t+1}} \sim \mathcal{N}(\mu = \hat{IP}, \sigma^2 = .25)$$

$$= P(\mathbf{OR_{t+1}}) = \frac{1}{.25\sqrt{2\pi}} e^{-\left(x-\hat{IP}\right)^2/2\times.25^2}$$

In order to discourage gaming of the system, we also draw from a normal distribution centered around 0 ~50% of the time.

$$\mathbf{OR_{t+1}} \sim \mathcal{N}(\mu = 0, \sigma^2 = .25)$$

$$= P(\mathbf{OR_{t+1}}) = \frac{1}{.25\sqrt{2\pi}} e^{-(x)^2/2\times.25^2}$$

Step 4: Randomly pick amounts to offer based on this ratio to present to the participant.

$$\mathbf{A_{Self,t+1}} \sim \mathcal{U}\left(\frac{.01}{min(1, OR_{t+1})}, \frac{10}{max(1, OR_{t+1})}\right)$$

$$\mathbf{A_{Other,t+1}} = A_S \times OR_{t+1}$$

Repeat this process with the next offer, $A_{Self,t+1}$ or $A_{Other,t+1}$.

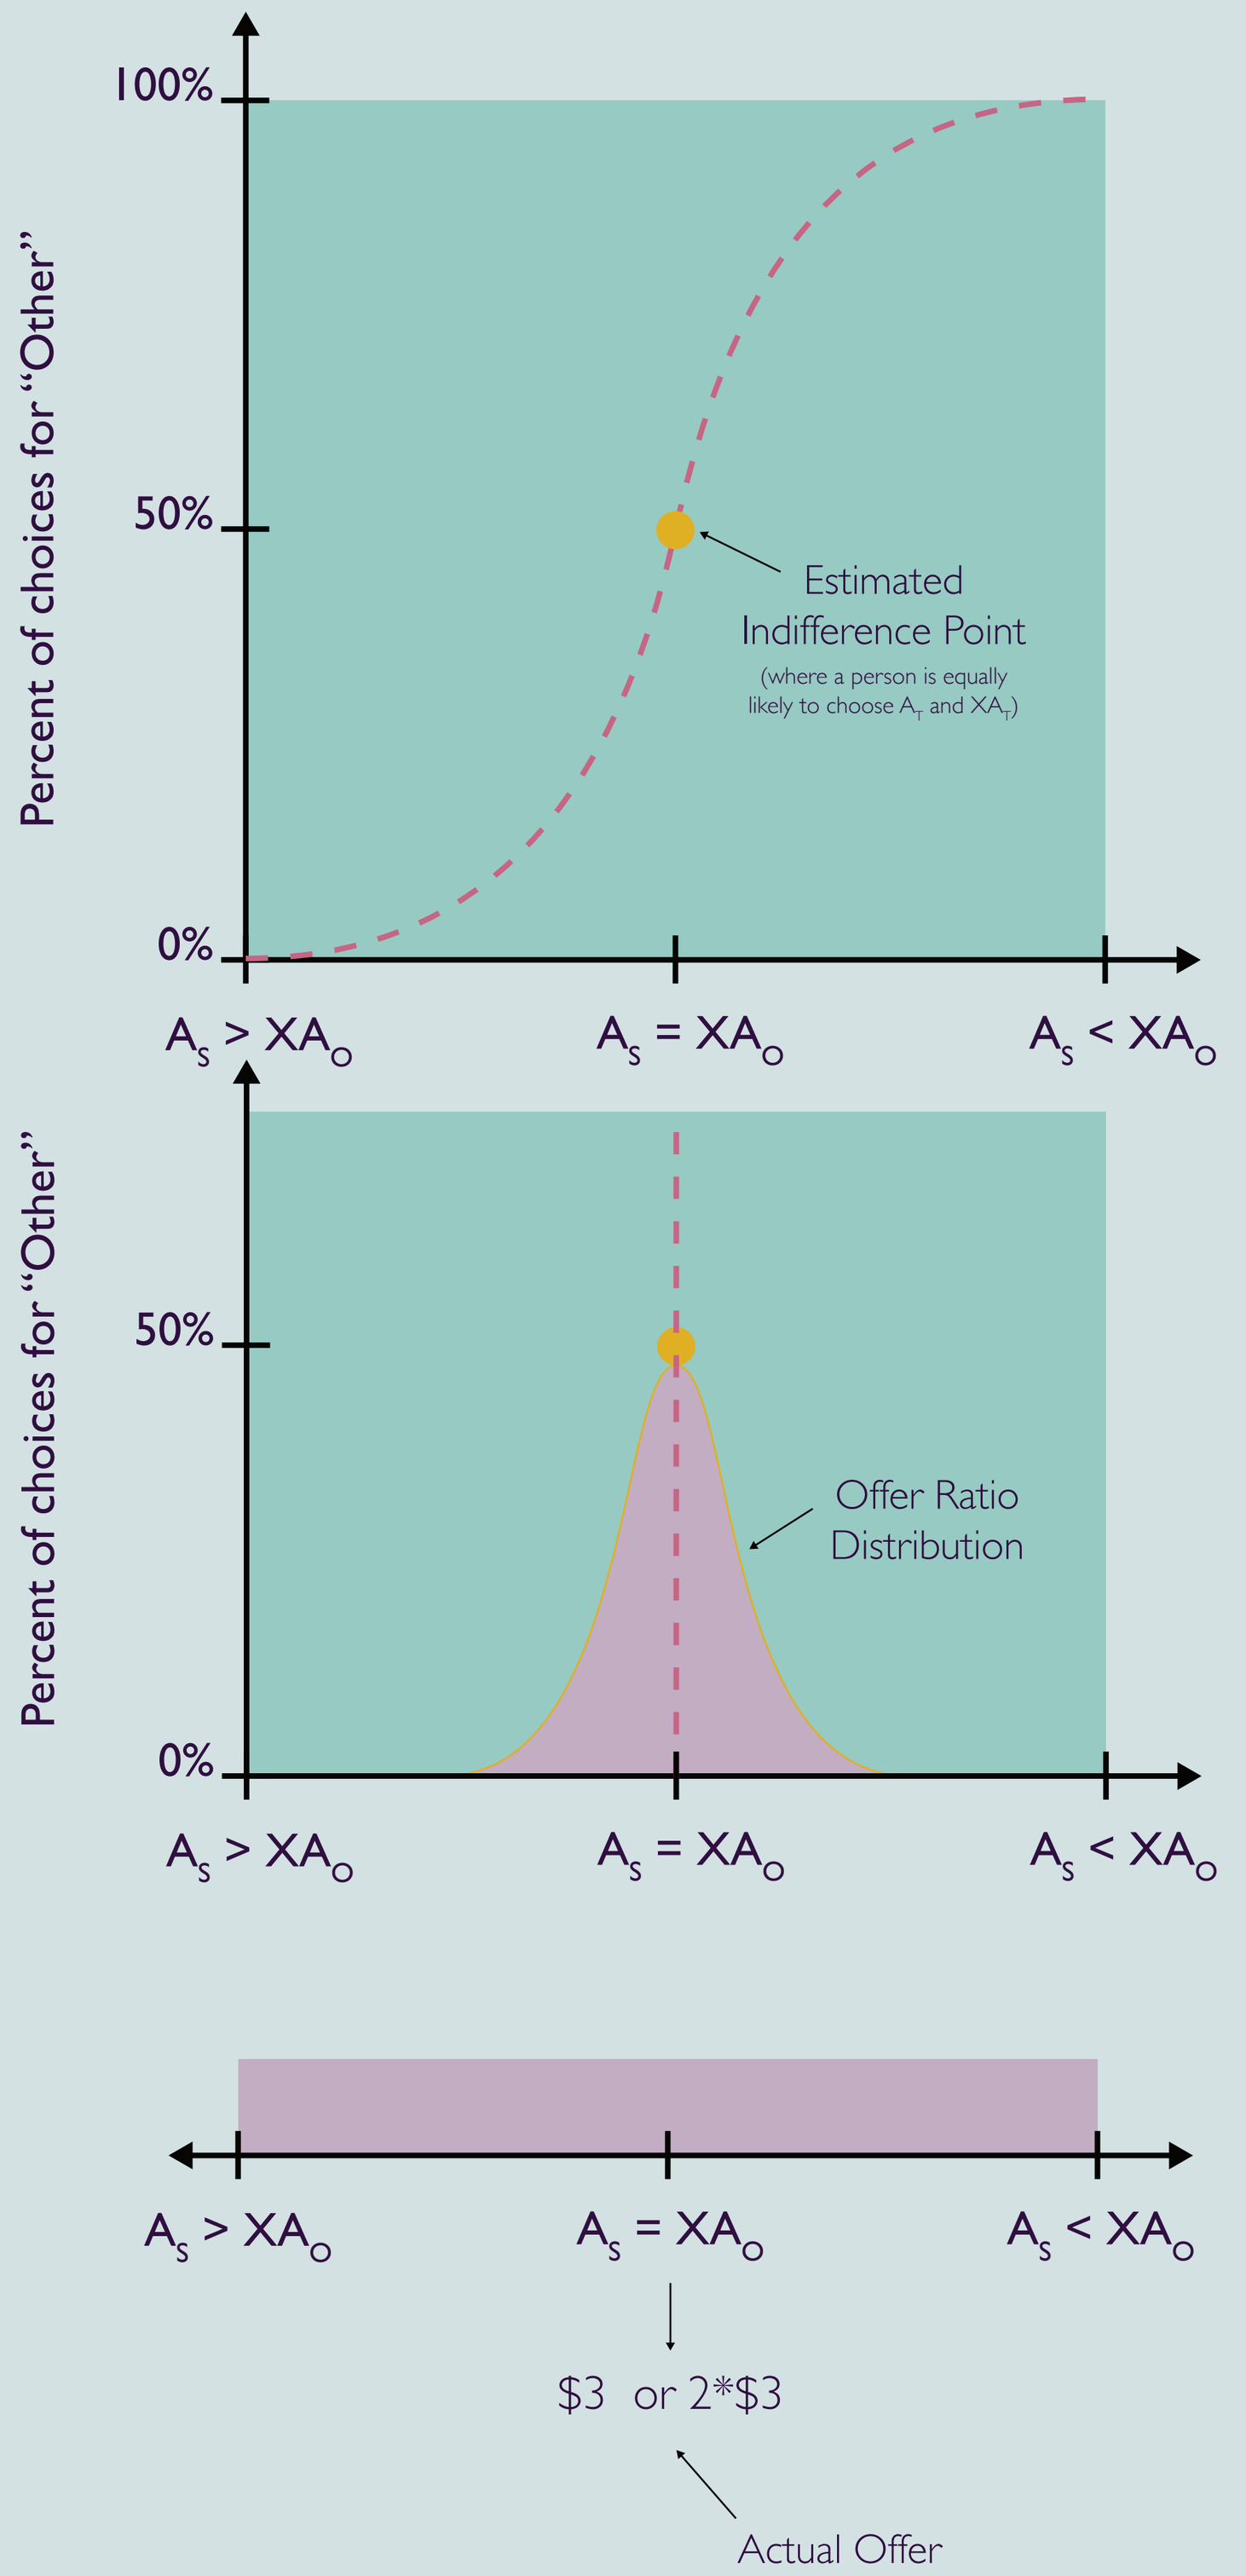
100%
50%
0%
Percent of choices for "Other"
Estimated Indifference Point
(where a person is equally likely to choose A_T and XA_T)
$A_s > XA_o$
$A_s = XA_o$
$A_s < XA_o$
Offer Ratio Distribution
$3 or 2*$3
Actual Offer

This study was funded by grants from the Fetzer Institute, The John Templeton Foundation, and NIMH MH43454 to RJD, and P30 HD008352. We would also like to thank Liz Vanderwerff for her critical assistance, and the staff at the Waisman Center for Brain Imaging and Behavior.

fMRI Experiment

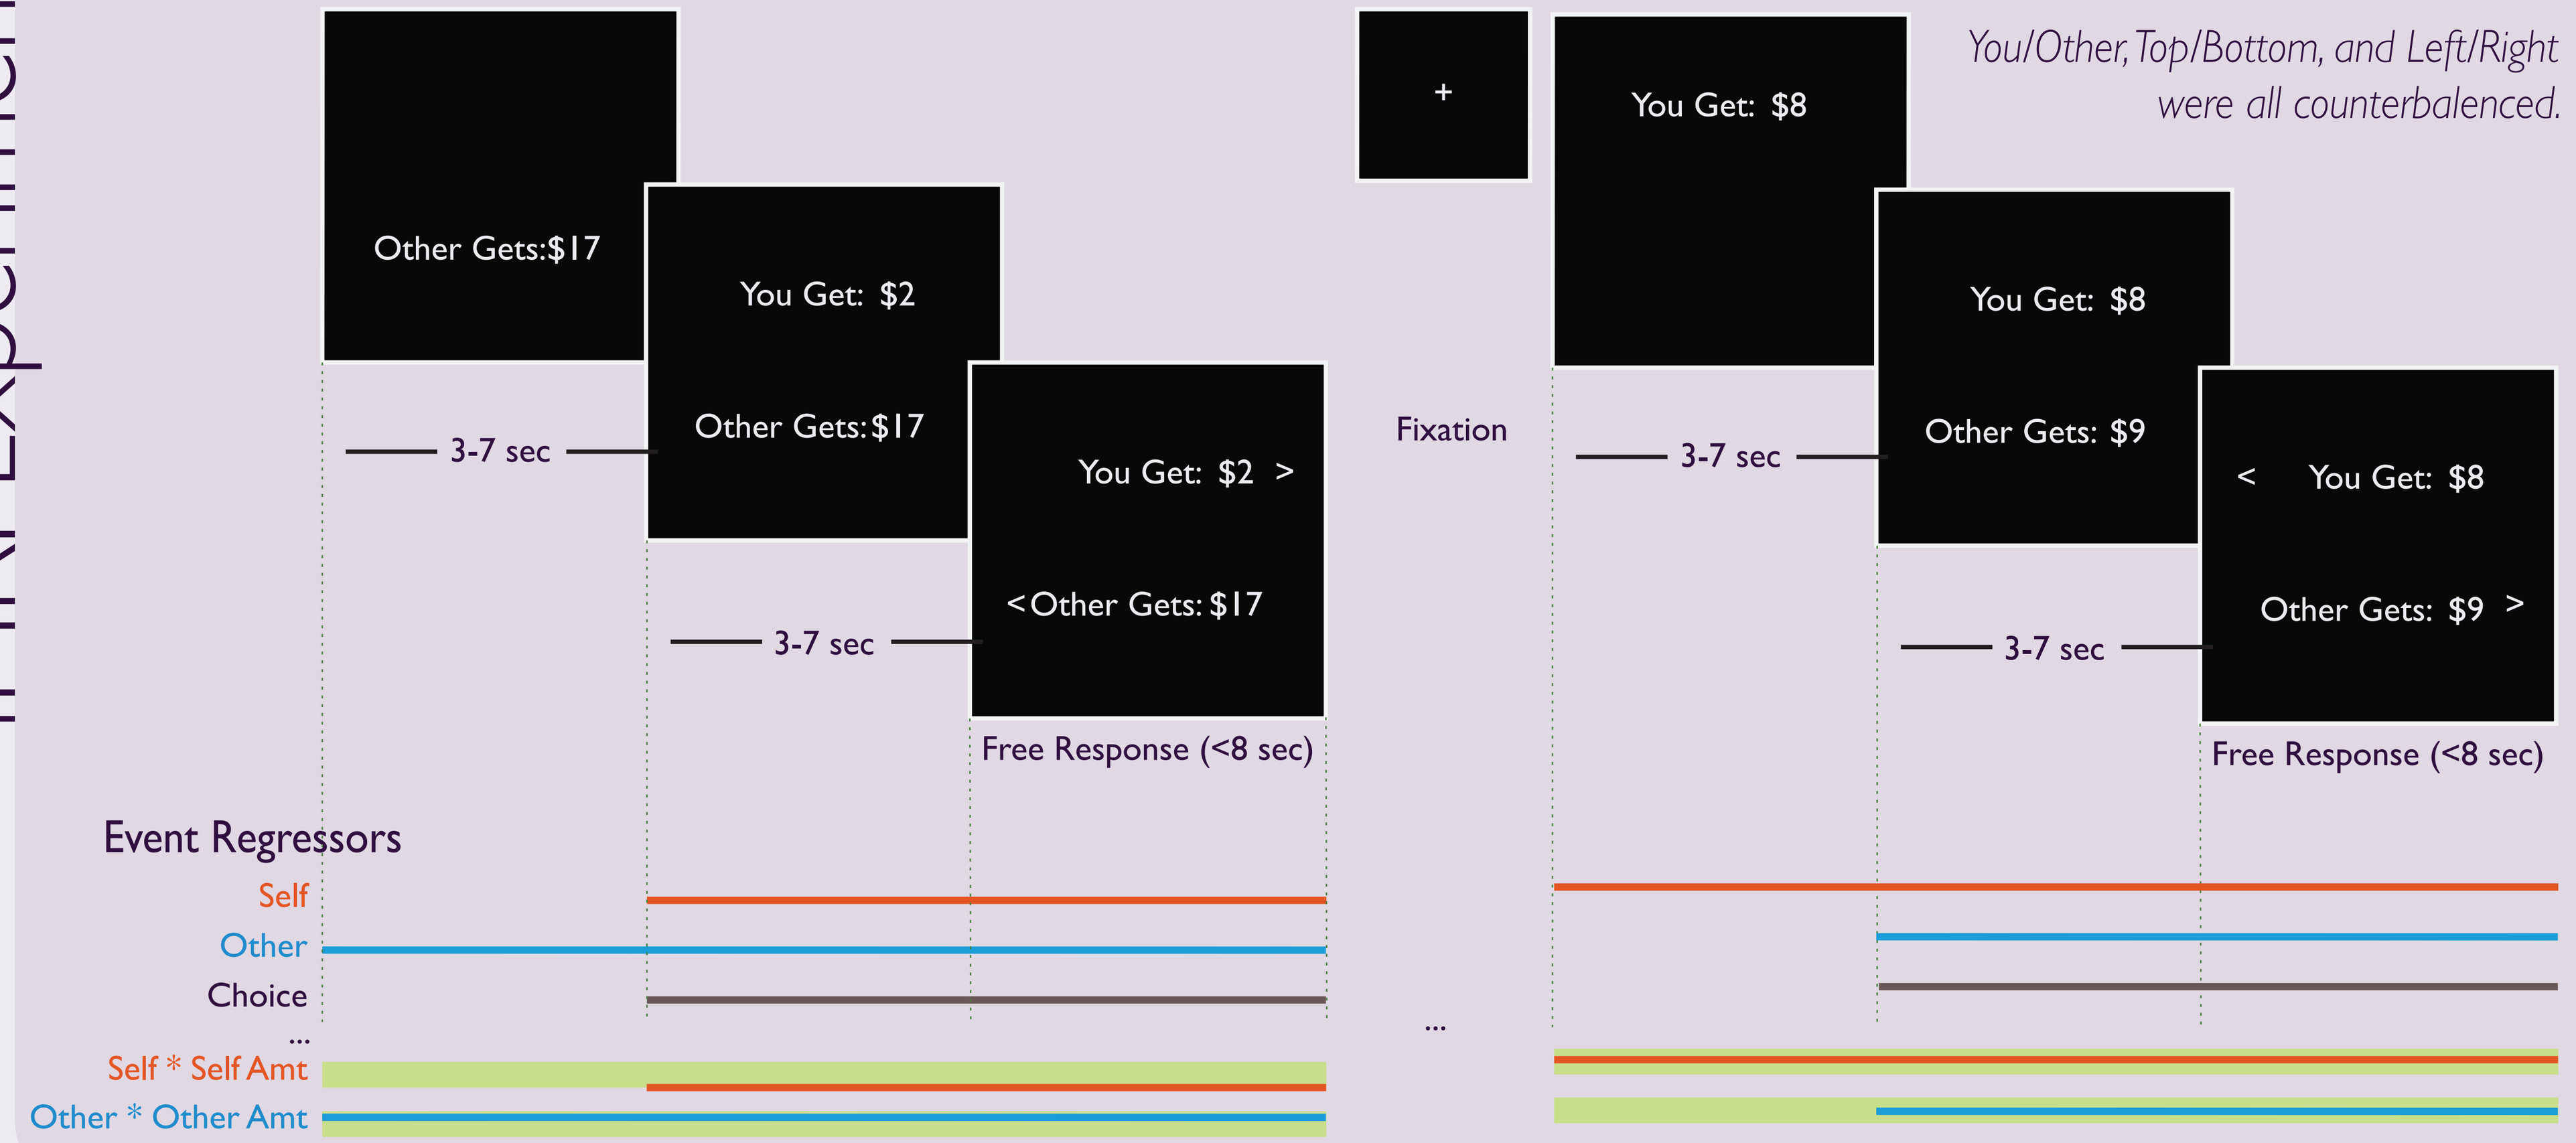
Other Gets:$17
You Get: $2
Other Gets:$17
You Get: $2 >
<Other Gets: $17
3-7 sec
Free Response (<8 sec)
+
Fixation
You Get: $8
You Get: $8
Other Gets: $9
< You Get: $8
Other Gets: $9 >
You/Other, Top/Bottom, and Left/Right were all counterbalenced.
Event Regressors
Self
Other
Choice
...
Self * Self Amt
Other * Other Amt

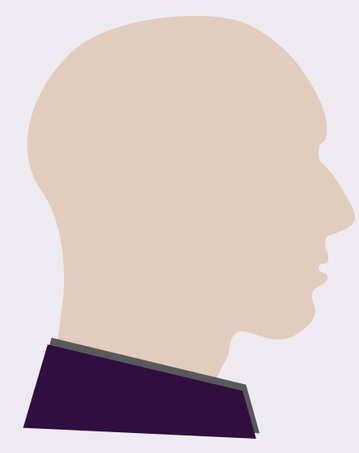

1) Subjects arrive and consent
2) 100 Trials of Self-Other in MRI
3) 200 trails of web-based IP estimation

In the fMRI task, 35 participants made 100 choices between some amount of money for themselves and some amount of money for another person. After the fMRI task, subjects made 200 choices on the web-based behavioral version of the task, seen to the left. Six subjects reported some amount of skepticism that the other participants were real, these subjects were excluded from all analyses, leaving 29 subjects. The amounts were determined based on our previous behavioral data collection, to present participants with the same offers, but maintain the ability to examine choices on each side of their indifference point. BOLD fMRI scans were acquired sagittally every 2.35 seconds (inplane: 1.72x1.72, 2mm slab skip 1mm). BOLD time-series images were motion corrected using FSL's mcflirt and smoothed using a 6mm gassian kernel. Eye-tracking data were acquired using Avotec goggles. Offers were presented as seen in the paradigm picture above. Self and other offer events were modeled when the offer was on the screen. Regressors were convolved with a canonical HRF and entered into a GLM. Within subject GLMs were performed using fMRIstat, using a design matrix adjusted to account for slice-time acquisitions. For each participant, parameter estimates for parametrically modulated self-amount and other-amount regressors, as well as their difference, were computed. Parameter estimates were rigid-body aligned to the structural T1-weighted anatomical. Structural scans were manually brain extracted and transformed to a standard space using ANTS, and transformations were applied to the parameter estimate maps. Outside of the scanner, each participant performed our RPP task online. Voxelwise across-subject analyses between individual participant parameter estimates and RPP self-other indifference points were performed in matlab using fMRIStat. Multiple comparison correction was performed using an intensity threshold of p<.005, two-tailed, and p<.05 cluster-size correction using non-isotropic random field theory. Because selfish subjects were outlying, and potentially using a different strategy, all individual difference correlations presented here are excluding participants with IP>2.

Self * Self Amt - Other * Other Amt = Greater Amount Tracking in Self
Other * Other Amt - Self * Self Amt = Greater Amount Tracking in Other

The vmPFC tracked self-amount more than other amount, on average.

p<.005, FWE Corrected p<.05
p<.005, FWE Corrected p<.05

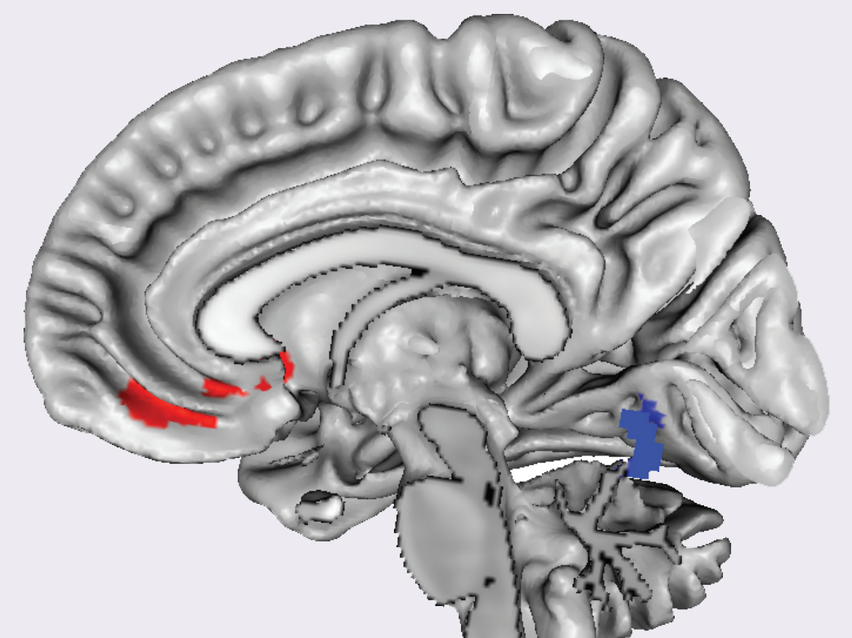

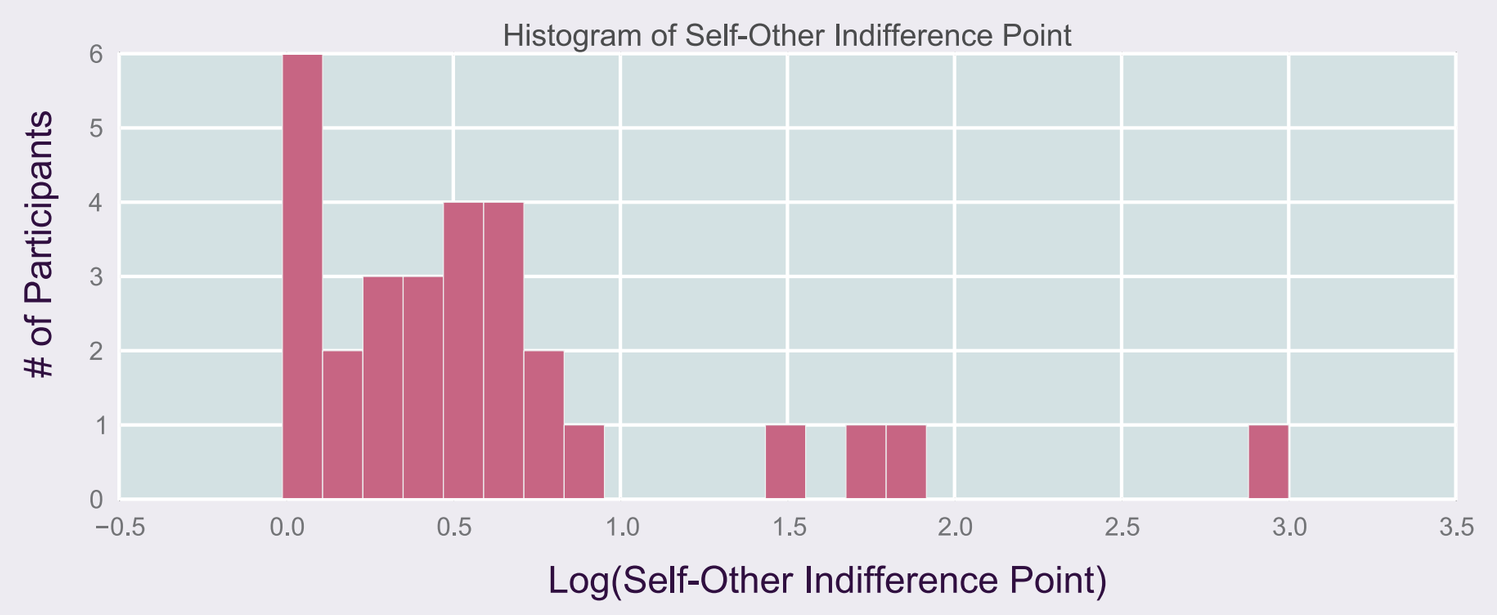
Histogram of Self-Other Indifference Point
of Participants
Log(Self-Other Indifference Point)

Behavioral Experiment

We tested 132 young adults using our adaptive online revealed prosocial preference task (RPP), in which participants made 200 choices between some amount of money for themselves and some amount of money for another person. There were substantial individual differences in self-other indifference point (IP) across participants (μ=.85, σ2=1.31). These data emphasize that different individuals have different pro-social preferences.

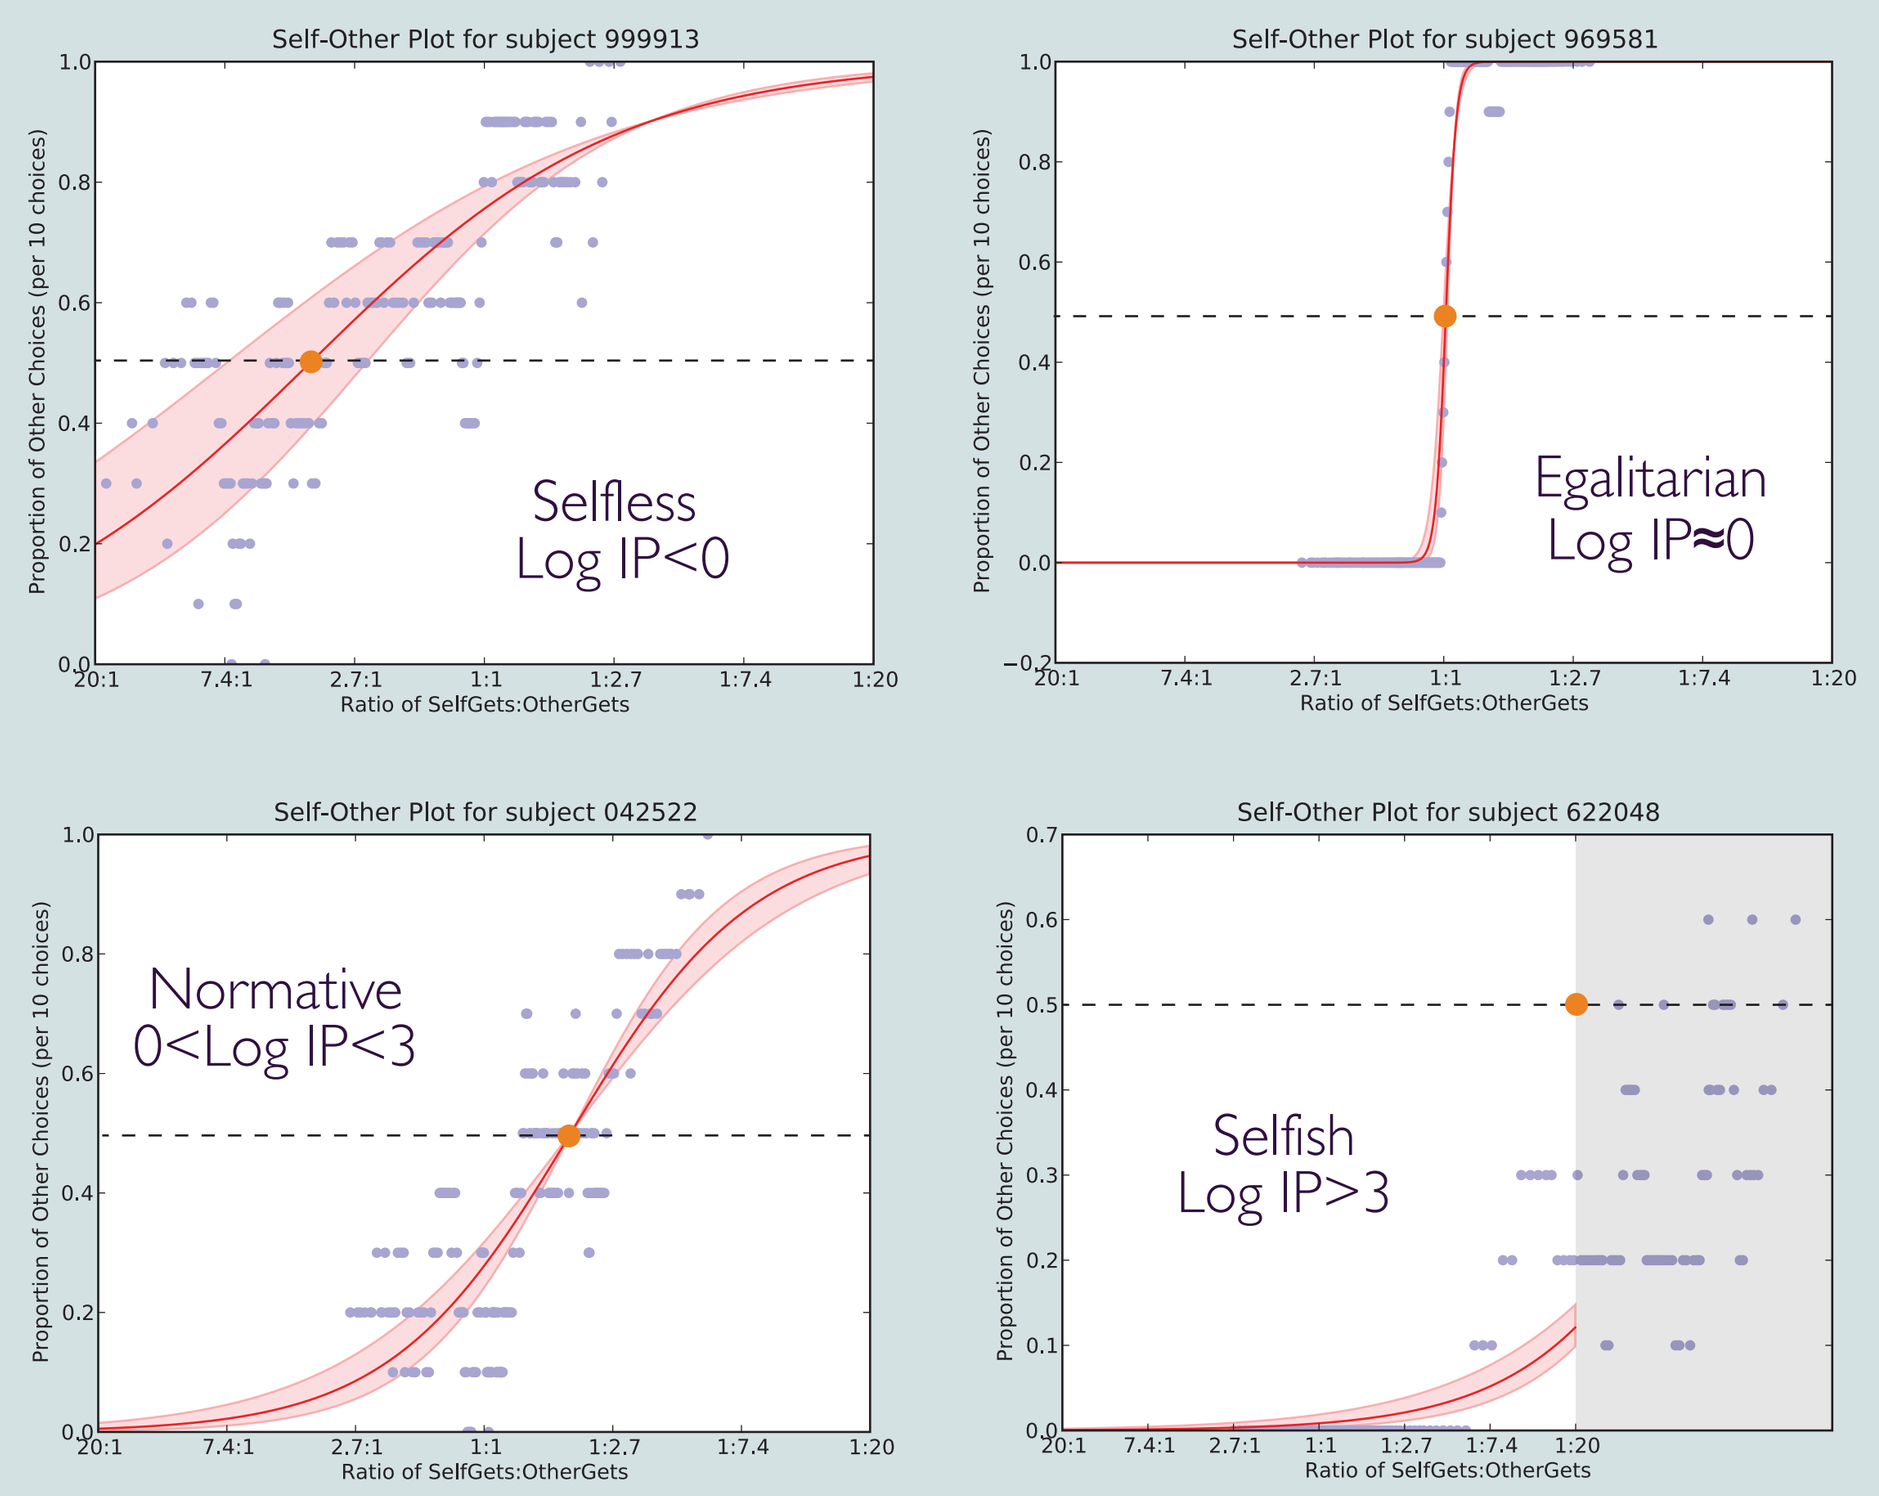
Self-Other Plot for subject 999913
Selfless
Log IP<0
Self-Other Plot for subject 969581
Egalitarian
Log IP≈0
Self-Other Plot for subject 042522
Normative
0<Log IP<3
Self-Other Plot for subject 622048
Selfish
Log IP>3
Proportion of Other Choices (per 10 choices)
Ratio of SelfGets:OtherGets

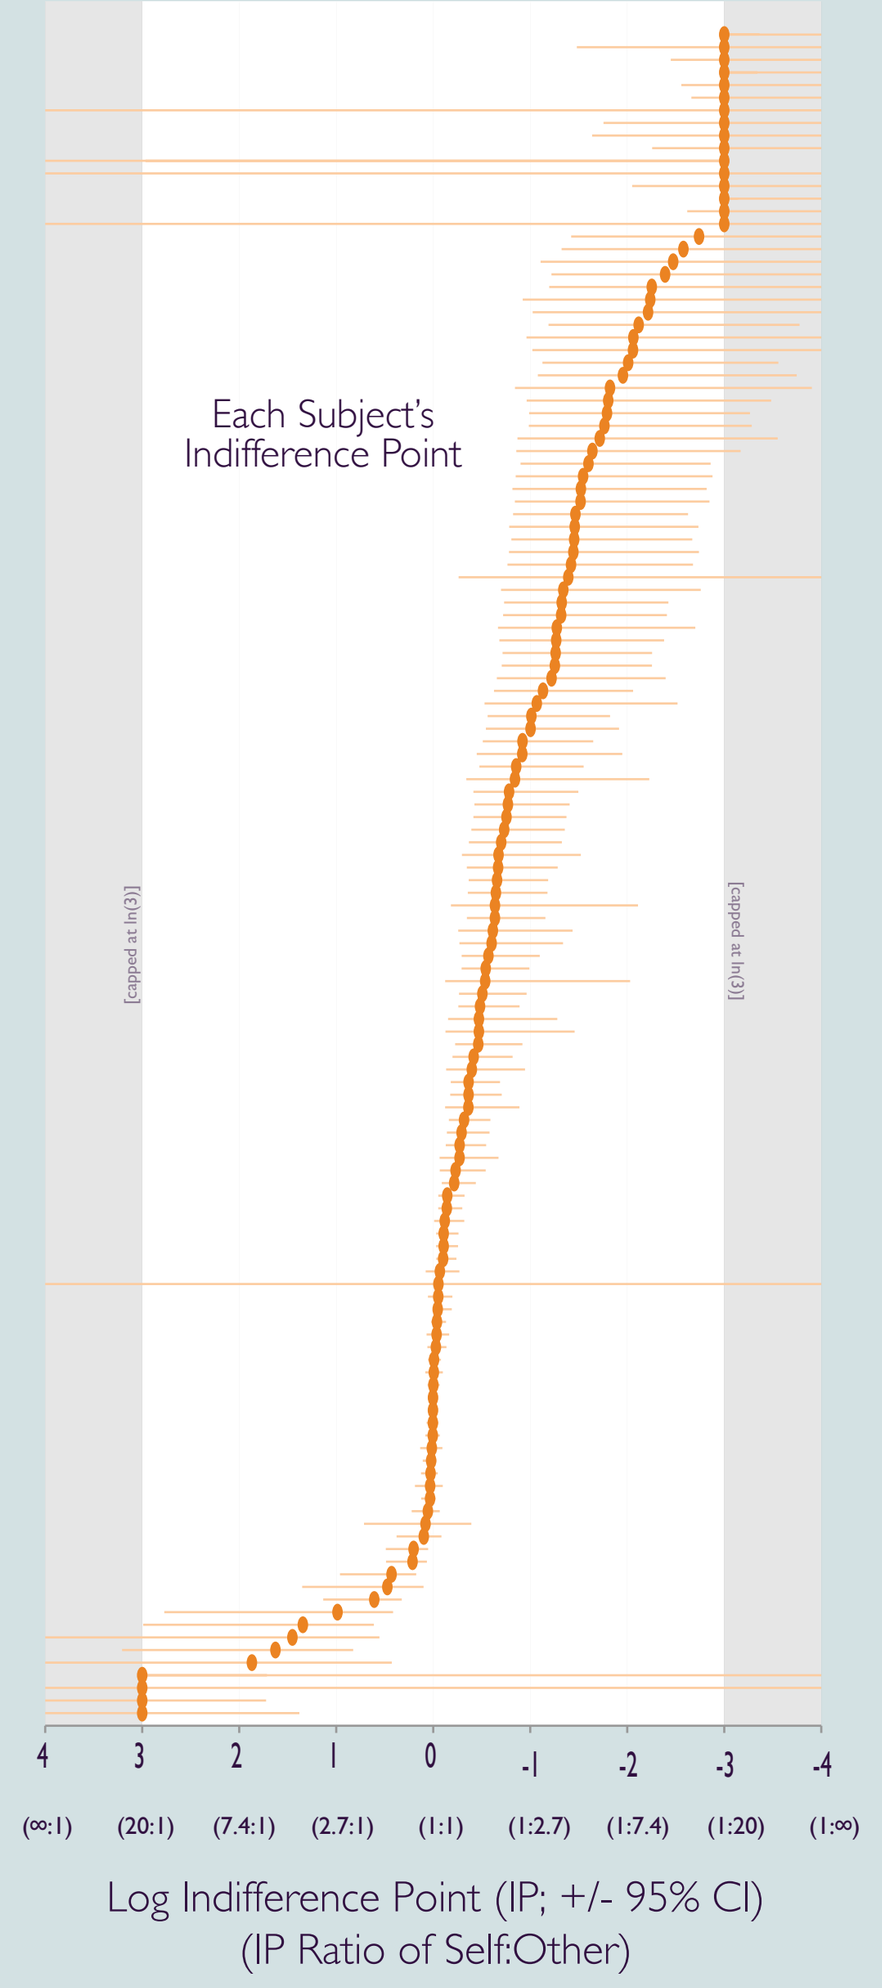
Each Subject's Indifference Point
Log Indifference Point (IP; +/- 95% CI)
(IP Ratio of Self:Other)

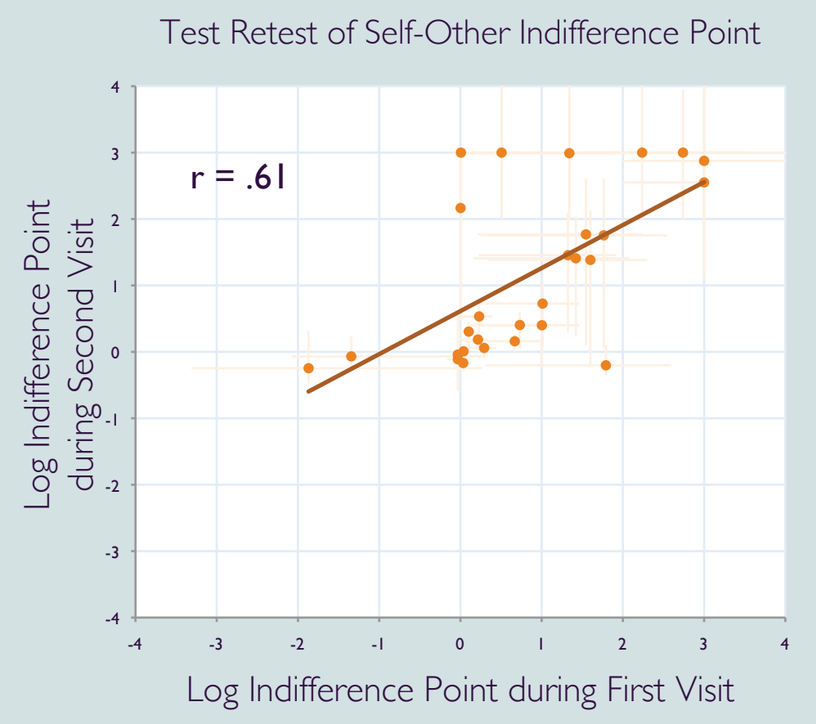
Test Retest of Self-Other Indifference Point
r = .61
Log Indifference Point during Second Visit
Log Indifference Point during First Visit

We examined the reliability of self-other IP as a measure of individual differences by testing 28 adults in the RPP two times, with 2 to 14 weeks between assessments. We observed significant correlations between IPs across time (r=.61, p=.0006; ICC=.61). These data suggest that individual differences in self-other IP are reliable over time and reflective of a trait-like tendency to value money to others.

Neural valuation coding of self amount vs. other amount predicts generosity

Self*Self Amt - Other*Other Amt = β_0*Constant + β_1*(Self-Other IP) + β_2*Age

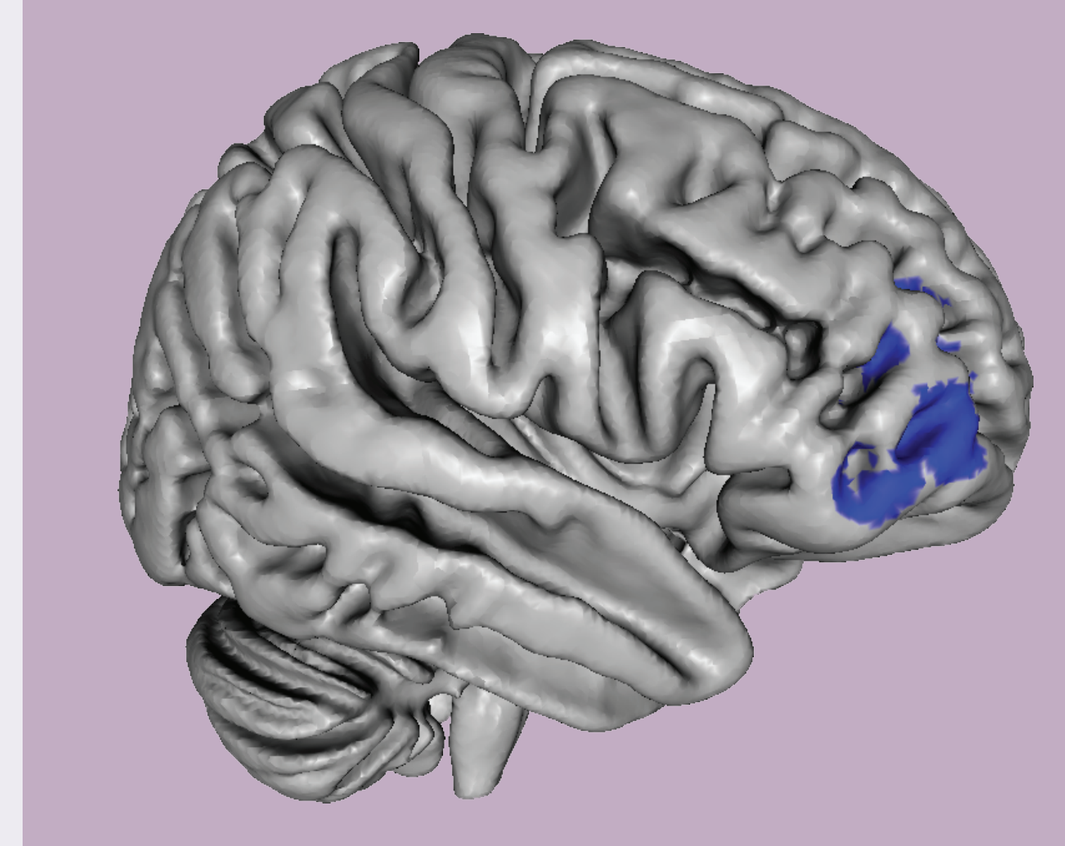

Lateral Prefrontal Cortex

The degree to which lateral prefrontal cortex tracks the value of self and other offer amounts, predicts individual differences in self-other indifference point.

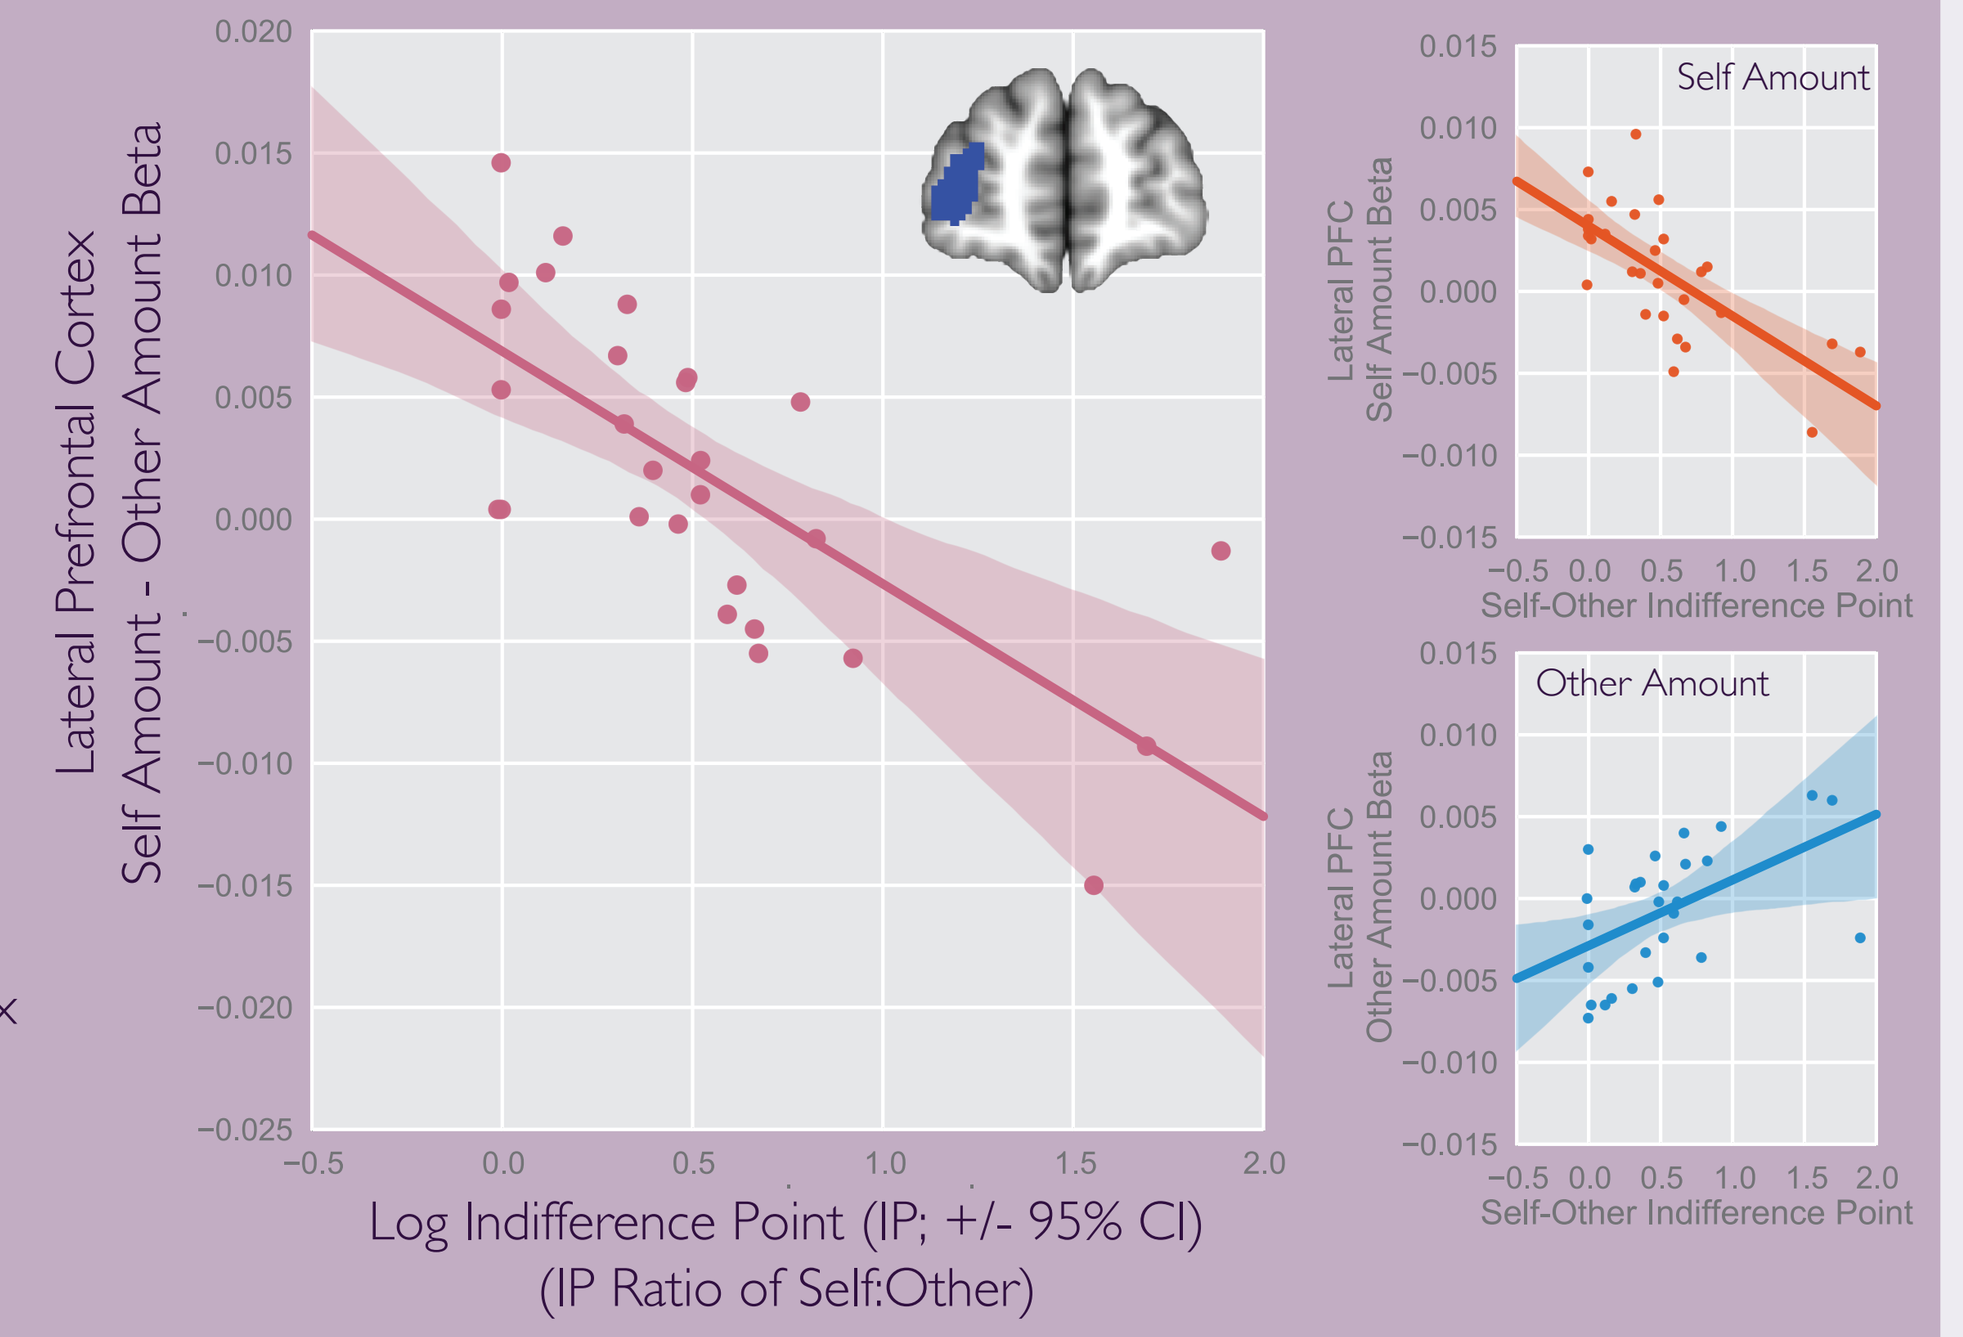
Lateral Prefrontal Cortex
Self Amount - Other Amount Beta
Log Indifference Point (IP; +/- 95% CI)
(IP Ratio of Self:Other)
Self Amount
Other Amount
Self-Other Indifference Point

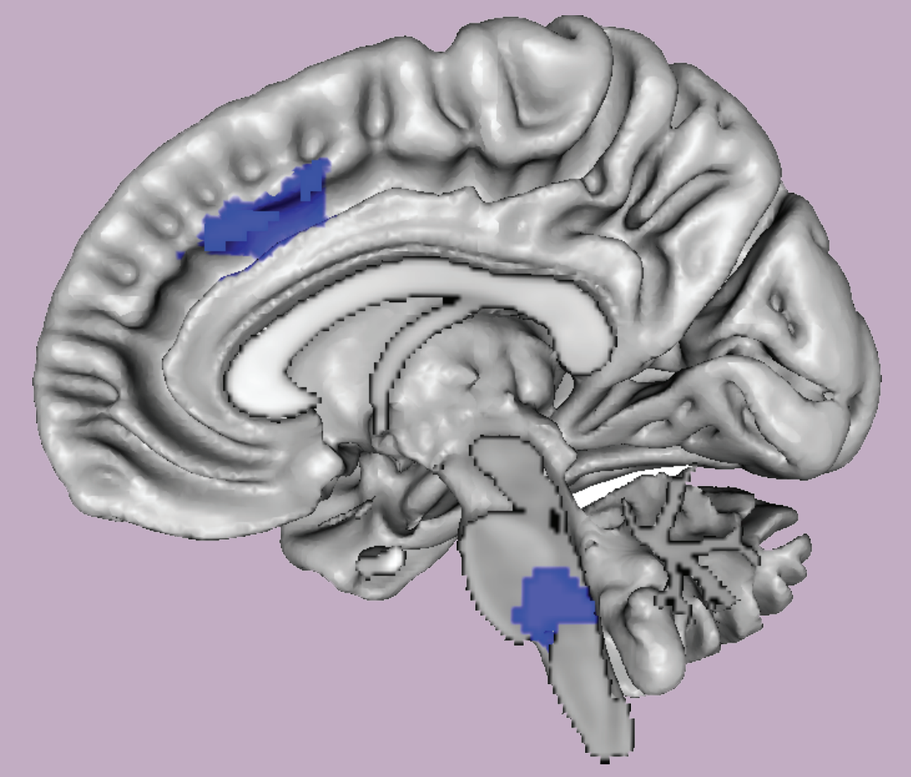

Anterior Mid-Cingulate

The degree to which anterior mid-cingulate cortex tracks the value of self and other offer amounts, predicts individual differences in self-other indifference point.

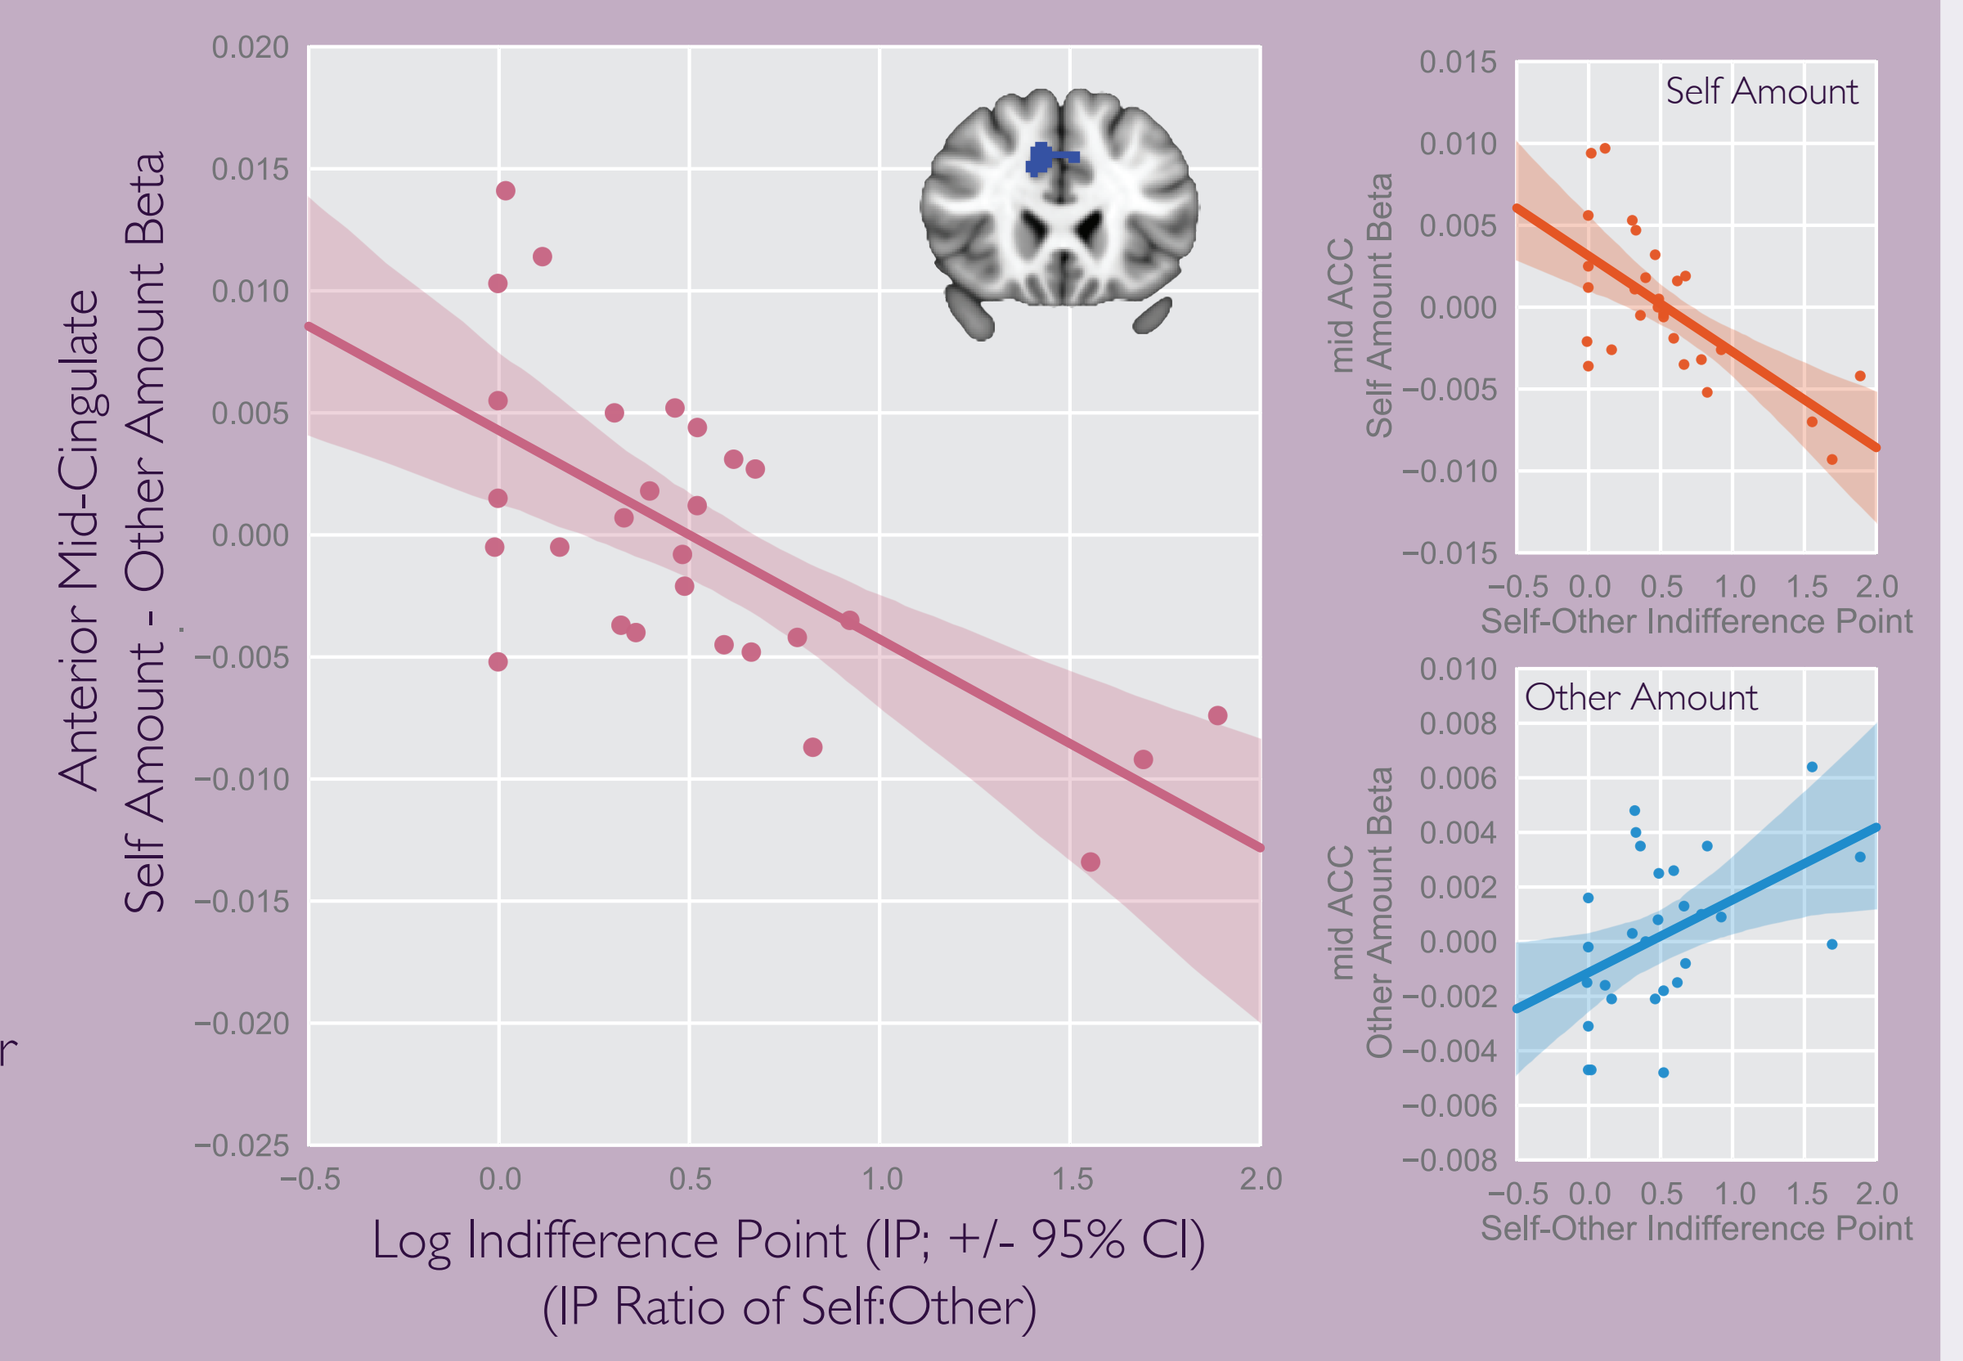
Anterior Mid-Cingulate
Self Amount - Other Amount Beta
Log Indifference Point (IP; +/- 95% CI)
(IP Ratio of Self:Other)
Self Amount
Other Amount
Self-Other Indifference Point

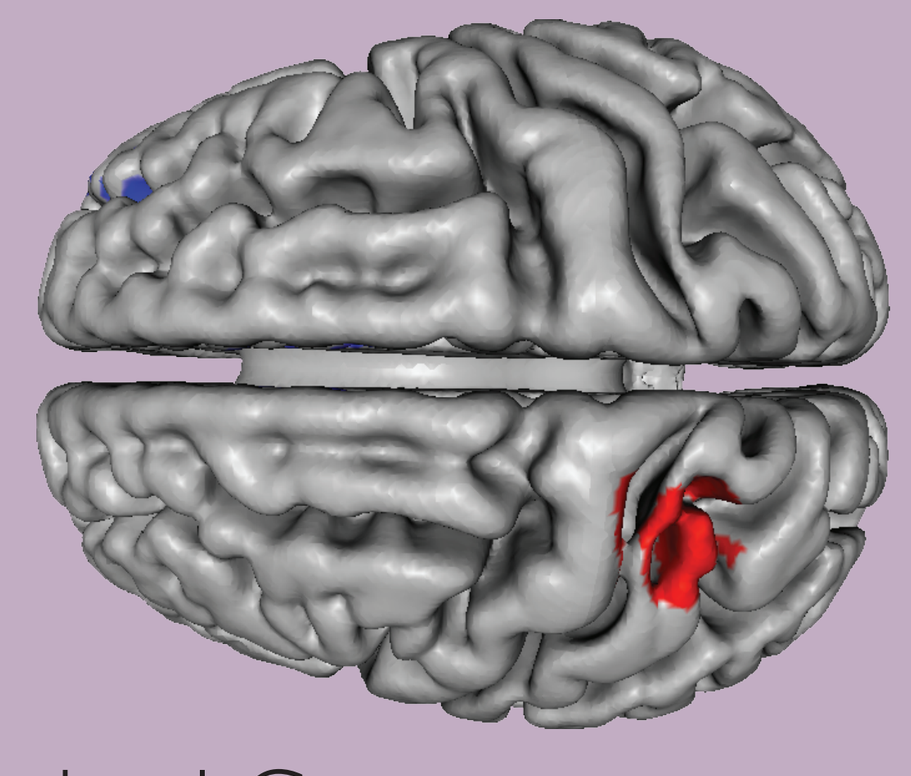

Parietal Cortex

The degree to which somatosensory cortex tracks the value of self and other offer amounts, predicts individual differences in self-other indifference point.

p<.005, FWE Corrected p<.05
p<.005, FWE Corrected p<.05

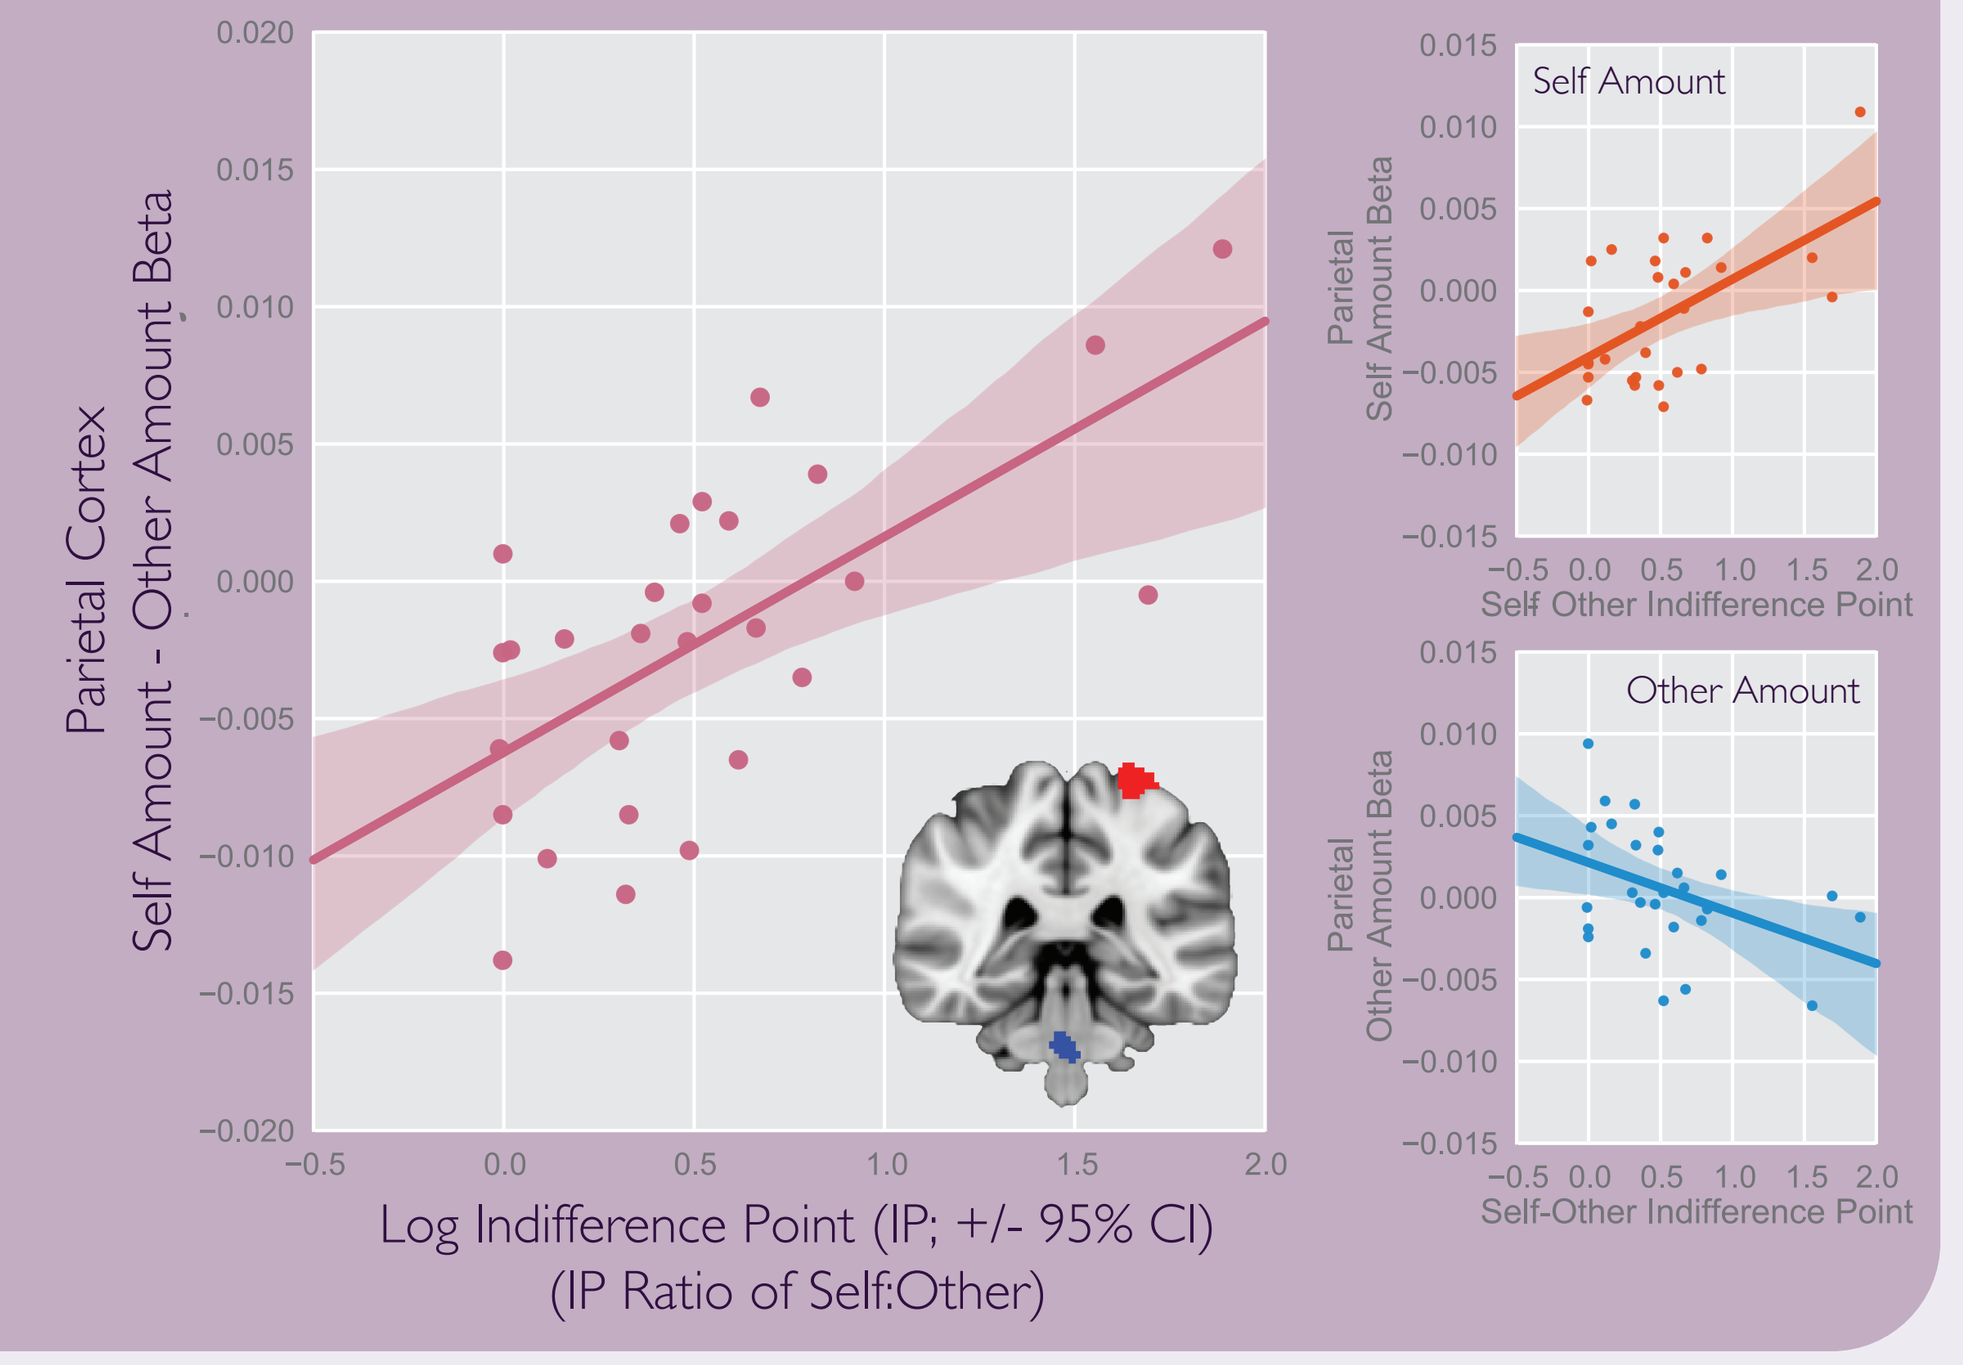
Parietal Cortex
Self Amount - Other Amount Beta
Log Indifference Point (IP; +/- 95% CI)
(IP Ratio of Self:Other)
Self Amount
Other Amount
Self-Other Indifference Point

We developed a reliable way to measure generosity using our RPP task, and demonstrated that individual differences in neural valuation signals are associated with variation in other-oriented preferences.

Try it yourself!
http://webtasks.keck.waisman.wisc.edu/self_other_demo